Lucerastat effect on kidney function in patients with Fabry Disease: Results from the ongoing open label-extension (OLE) of the phase 3 clinical program

Eric Wallace[1], Dominique P. Germain[2,3], Peter Blattmann[4], Aline Frey[4], Kolbeinn Gudmundsson[4], Derralynn Hughes[5,6], Michael Meinel[4], Peter Nordbeck[7,8], Christoph Wanner[8]

[1]University of Alabama at Birmingham, Birmingham, AL, United States; [2]French Referral Centre for Fabry Disease and Lysosomal disorders, MetabERN European Reference Network, Paris Saclay University Paris, France; [3]Division of Medical Genetics, University of Versailles, Montigny, France; [4]Idorsia Pharmaceuticals Ltd., Allschwil, Basel-Landschaft, Switzerland; [5]University College London, London, United Kingdom; [6]Royal Free London NHS Foundation Trust, London, United Kingdom; [7]Department of Internal Medicine I, University Hospital Würzburg, Würzburg, Germany; 8Fabry Center for Interdisciplinary Therapy (FAZiT), University Hospital Würzburg, Würzburg, Germany;

Background

- Fabry disease (FD) is a rare, X-linked, lysosomal storage disorder caused by pathogenic variants in the *GLA* gene, which encodes the α-galactosidase A (α-Gal A) enzyme.[1]
- Deficient α-Gal A activity causes progressive and toxic accumulation of glycosphingolipids, including globotriaosylceramide (Gb3), in lysosomes, leading to organ dysfunction and decreased quality of life.[1-3] Kidney involvement is one of the hallmark consequences of FD, with progressive Gb3 accumulation leading to proteinuria, declining eGFR and kidney damage.[1]
- Lucerastat, an oral glucosylceramide synthase (GCS) inhibitor, provides substrate reduction therapy by lowering Gb3 synthesis and limiting its lysosomal accumulation in multiple cell types in FD. We present 48-month interim findings from the ongoing global Phase 3 MODIFY open-label extension (OLE) study.[4,5]

Methods

- MODIFY[4] (NCT03425539) randomized adult patients with FD 2:1 to lucerastat 1000 mg b.i.d. orally (dose adjusted by eGFR) or placebo for 6 months. Participants who completed MODIFY were eligible to enter the OLE (NCT03737214) for up to 96 months, during which all received lucerastat.
- Prespecified efficacy endpoints included change from baseline in plasma Gb3 and annualized eGFR slope (mL/min/1.73m² per year) in the OLE. Analyses included participants with ≥2 pre-randomization creatinine values to derive a historical eGFR slope for comparison with the on-treatment slope.

Aim

The objective of this analysis was to evaluate the long-term effect of lucerastat on the renal function of subjects with FD and on the biomarkers of FD.

Results

Lucerastat slowed the decline of kidney function

- At Month-48, the lucerastat group exhibited reduced decline in the kidney function compared to historical controls in the overall population (eGFR slope, historical: -3.50; Lucerastat: -1.64). Efficacy was independent of gene variants.
- This effect was also seen in all subgroups analyzed, with the most prominent effects in subgroups with severe disease course, such as:
 - classic males (historical: -4.32; Lucerastat: -2.05),
 - patients with impaired renal function at baseline (historical: -6.18; Lucerastat: -2.49),
 - Anti-Drug Antibody (ADA+) positive patients (historical: -7.75; Lucerastat: -3.43)

Subgroup		Number of participants	eGFR slope (mL/min/1.73m2 per year)	LS Mean [95% CI]
Overall	Historical	93		-3.50 (-5.04, -1.96)
	Lucerastat	93		-1.64 (-2.33, -0.95)
Classic Fabry disease subtype by sex				
Classic female	Historical	36		-3.19 (-5.71, -0.67)
	Lucerastat	36		-1.21 (-2.39, -0.03)
Classic male	Historical	43		-4.32 (-6.59, -2.06)
	Lucerastat	43		-2.05 (-3.16, -0.94)
eGFR at baseline				
<90 mL/min/1.73m²	Historical	30		-6.18 (-8.68, -3.69)
	Lucerastat	30		-2.49 (-3.69, -1.28)
≥90 mL/min/1.73m²	Historical	63		-2.20 (-4.07, -0.33)
	Lucerastat	63		-1.22 (-2.03, -0.40)
ADA Status at 301 Baseline				
ADA+	Historical	16		-7.75 (-11.28, -4.21)
	Lucerastat	16		-3.43 (-5.14, -1.73)
ADA-	Historical	17		-1.76 (-5.07, 1.55)
	Lucerastat	17		-1.20 (-2.82, 0.42)

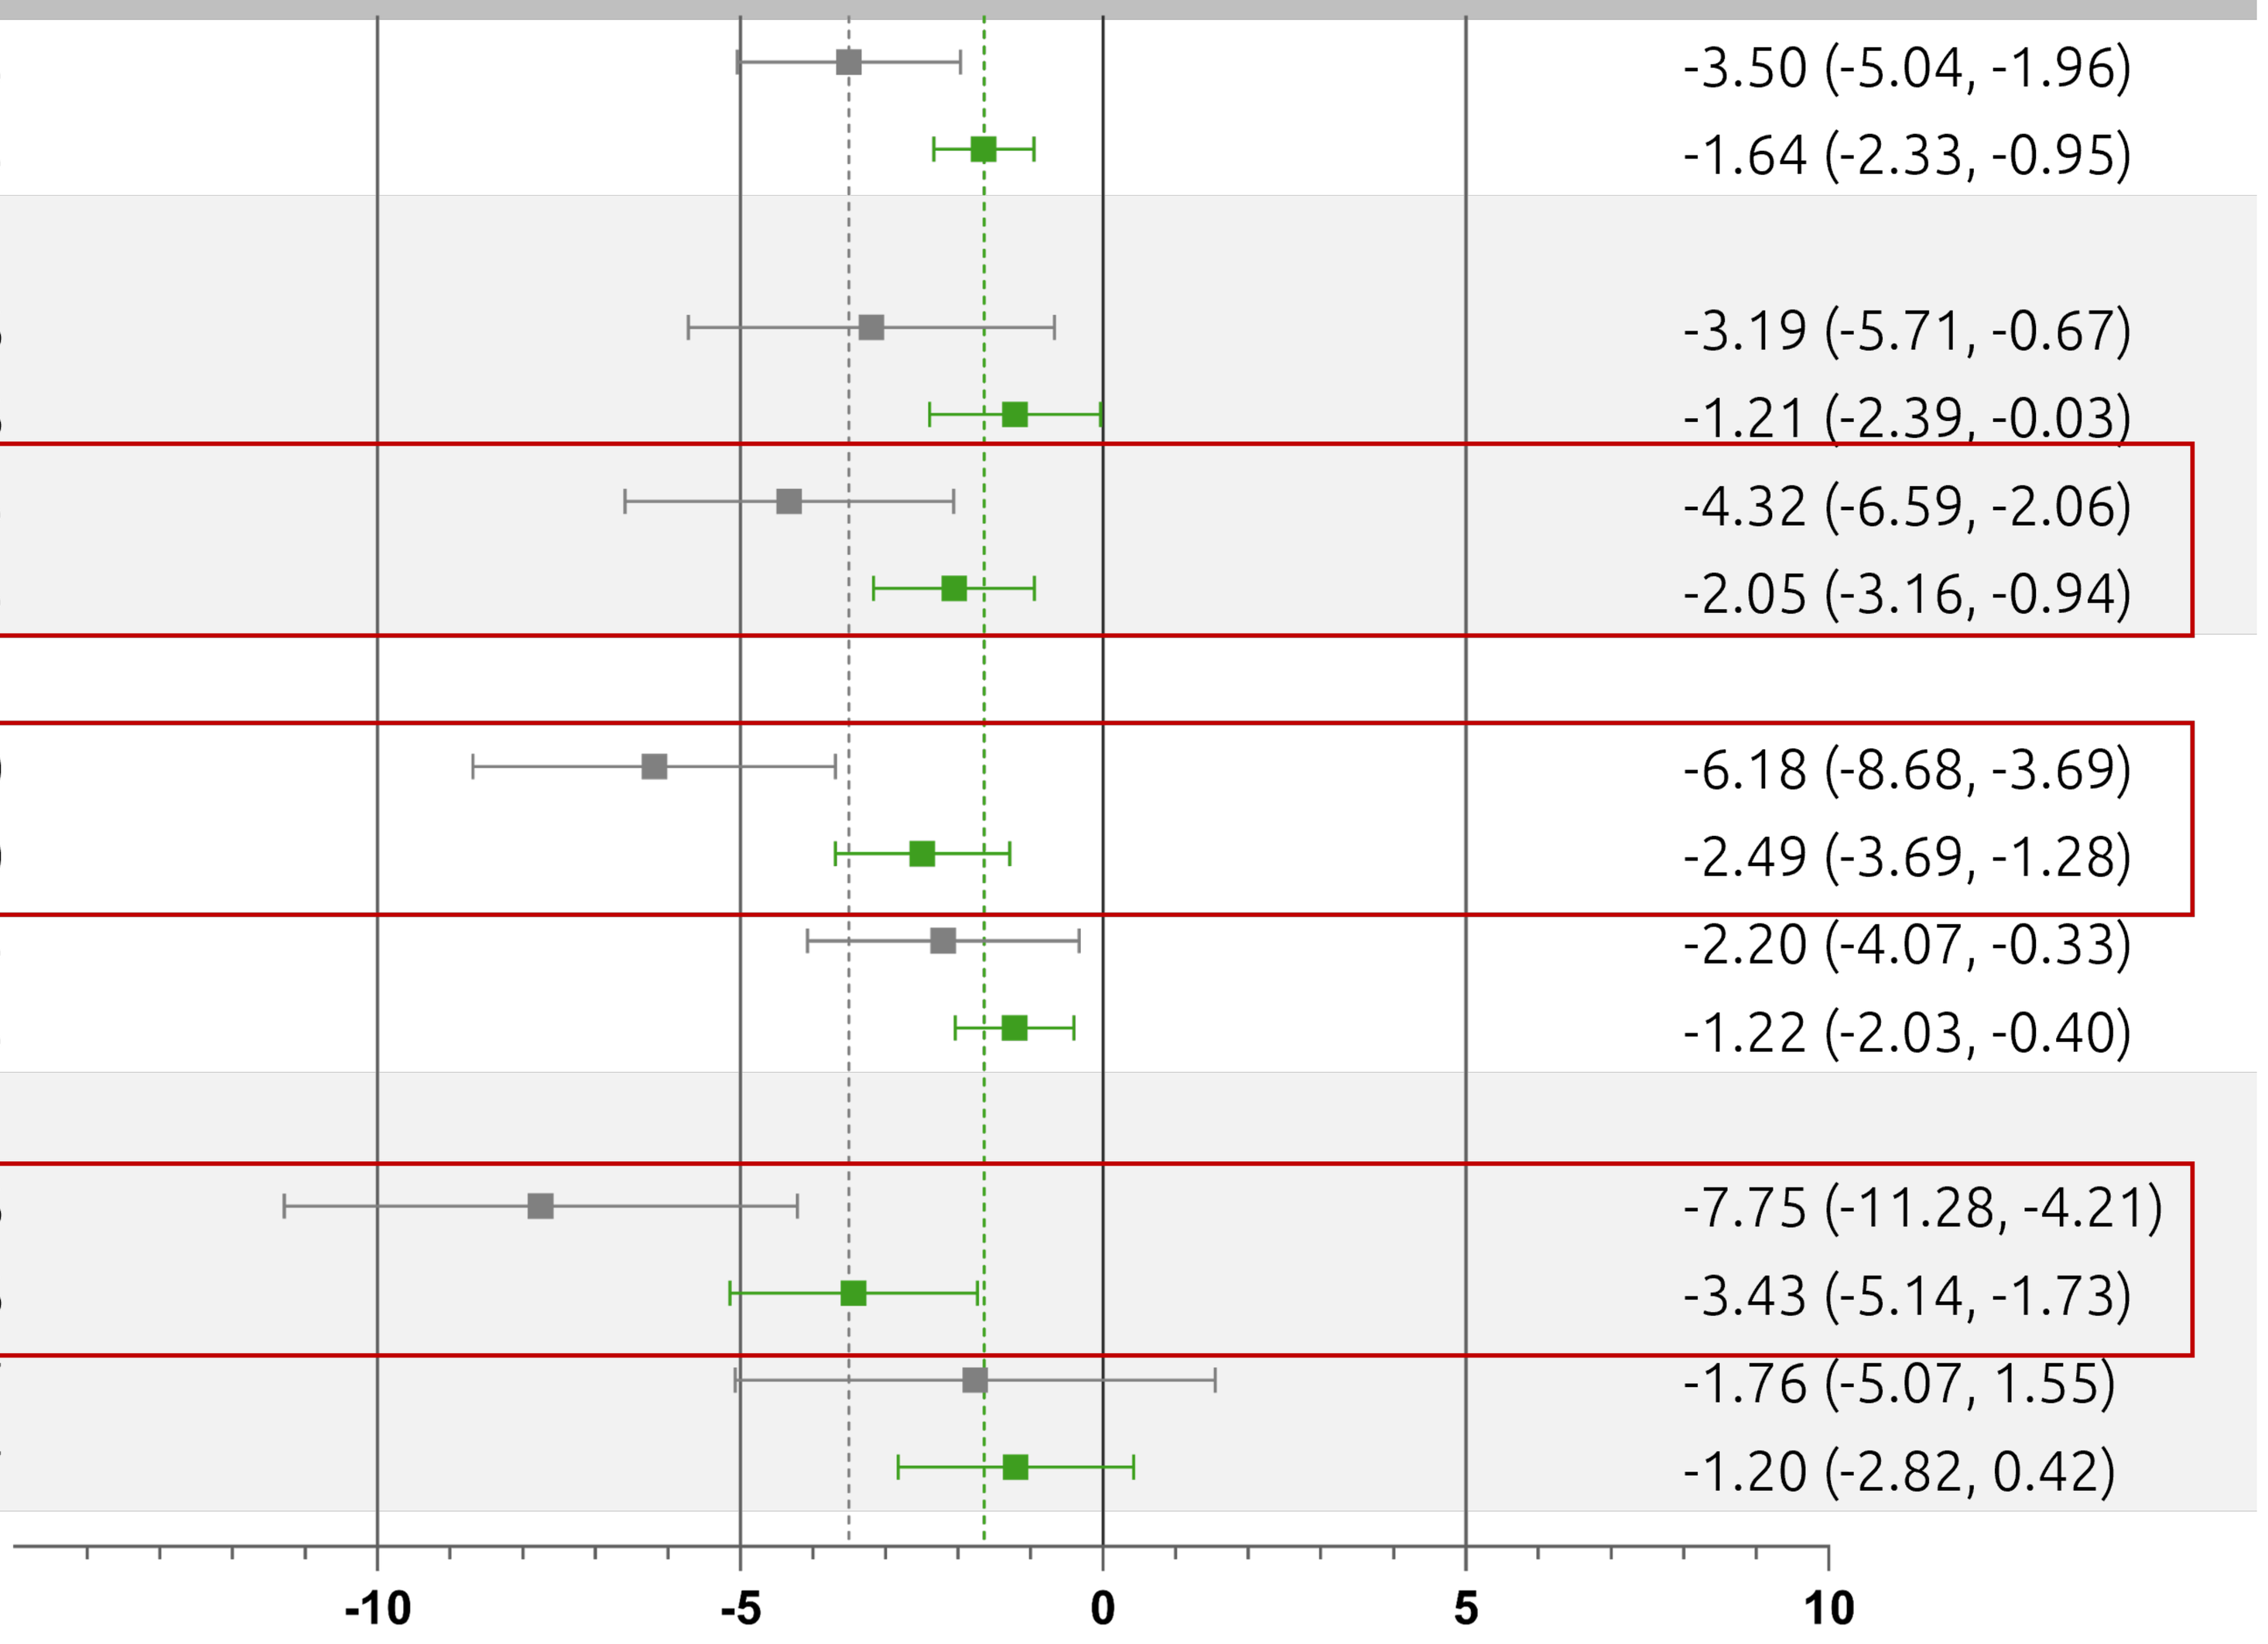
-10
-5
0
5
10

Baseline* demographics and disease characteristics

- Of 118 participants randomized, 117 received study treatment (80 lucerastat, 37 prior placebo) in MODIFY.
- 107 (91%) participants were enrolled in the OLE, and 55 (47%) ongoing at Month-48 OLE interim analysis.
- Participants' demographic and baseline characteristics were similar across initial treatment groups.

	Toral Lucerastat (N=118)
Age at screening, years, mean (SD)	39.3 (14.1)
Male, n (%)	55 (47%)
Race , n (%)	
White	113 (96%)
Asian	3 (3%)
Plasma Gb3 (ng/ml) , Mean (SD)	1702 (991)
Plasma LysoGb3 (ng/ml), Mean (SD)	32 (41)
Normalized urine Gb3 (µmol Gb3/mol creatinine), mean (SD)	9.9 (17.0)
ADA Status [n(%)]	
ADA+	18 (48.6)
ADA-	19 (51.4)
Missing	77
eGFR (mL/min/1.73m2), Mean (SD)	98.2 (25.2)
eGFR, ml/min/1.73m², n (%)	
≥90	77 (65%)
<90	41 (34%)
UACR (mg/g) , median (Q1, Q3)	23.0 (12.0, 114.8)
UACR, n (%)	
≥30	48 (42.1)
<30	66 (57.9)

Safety

Lucerastat was generally well-tolerated. No new safety signals were identified during the OLE as compared to MODIFY.

Lucerastat led to significant reduction in plasma and urine Gb3 levels

- Lucerastat led to a sustained 58% decrease in plasma Gb3 levels by Month 48

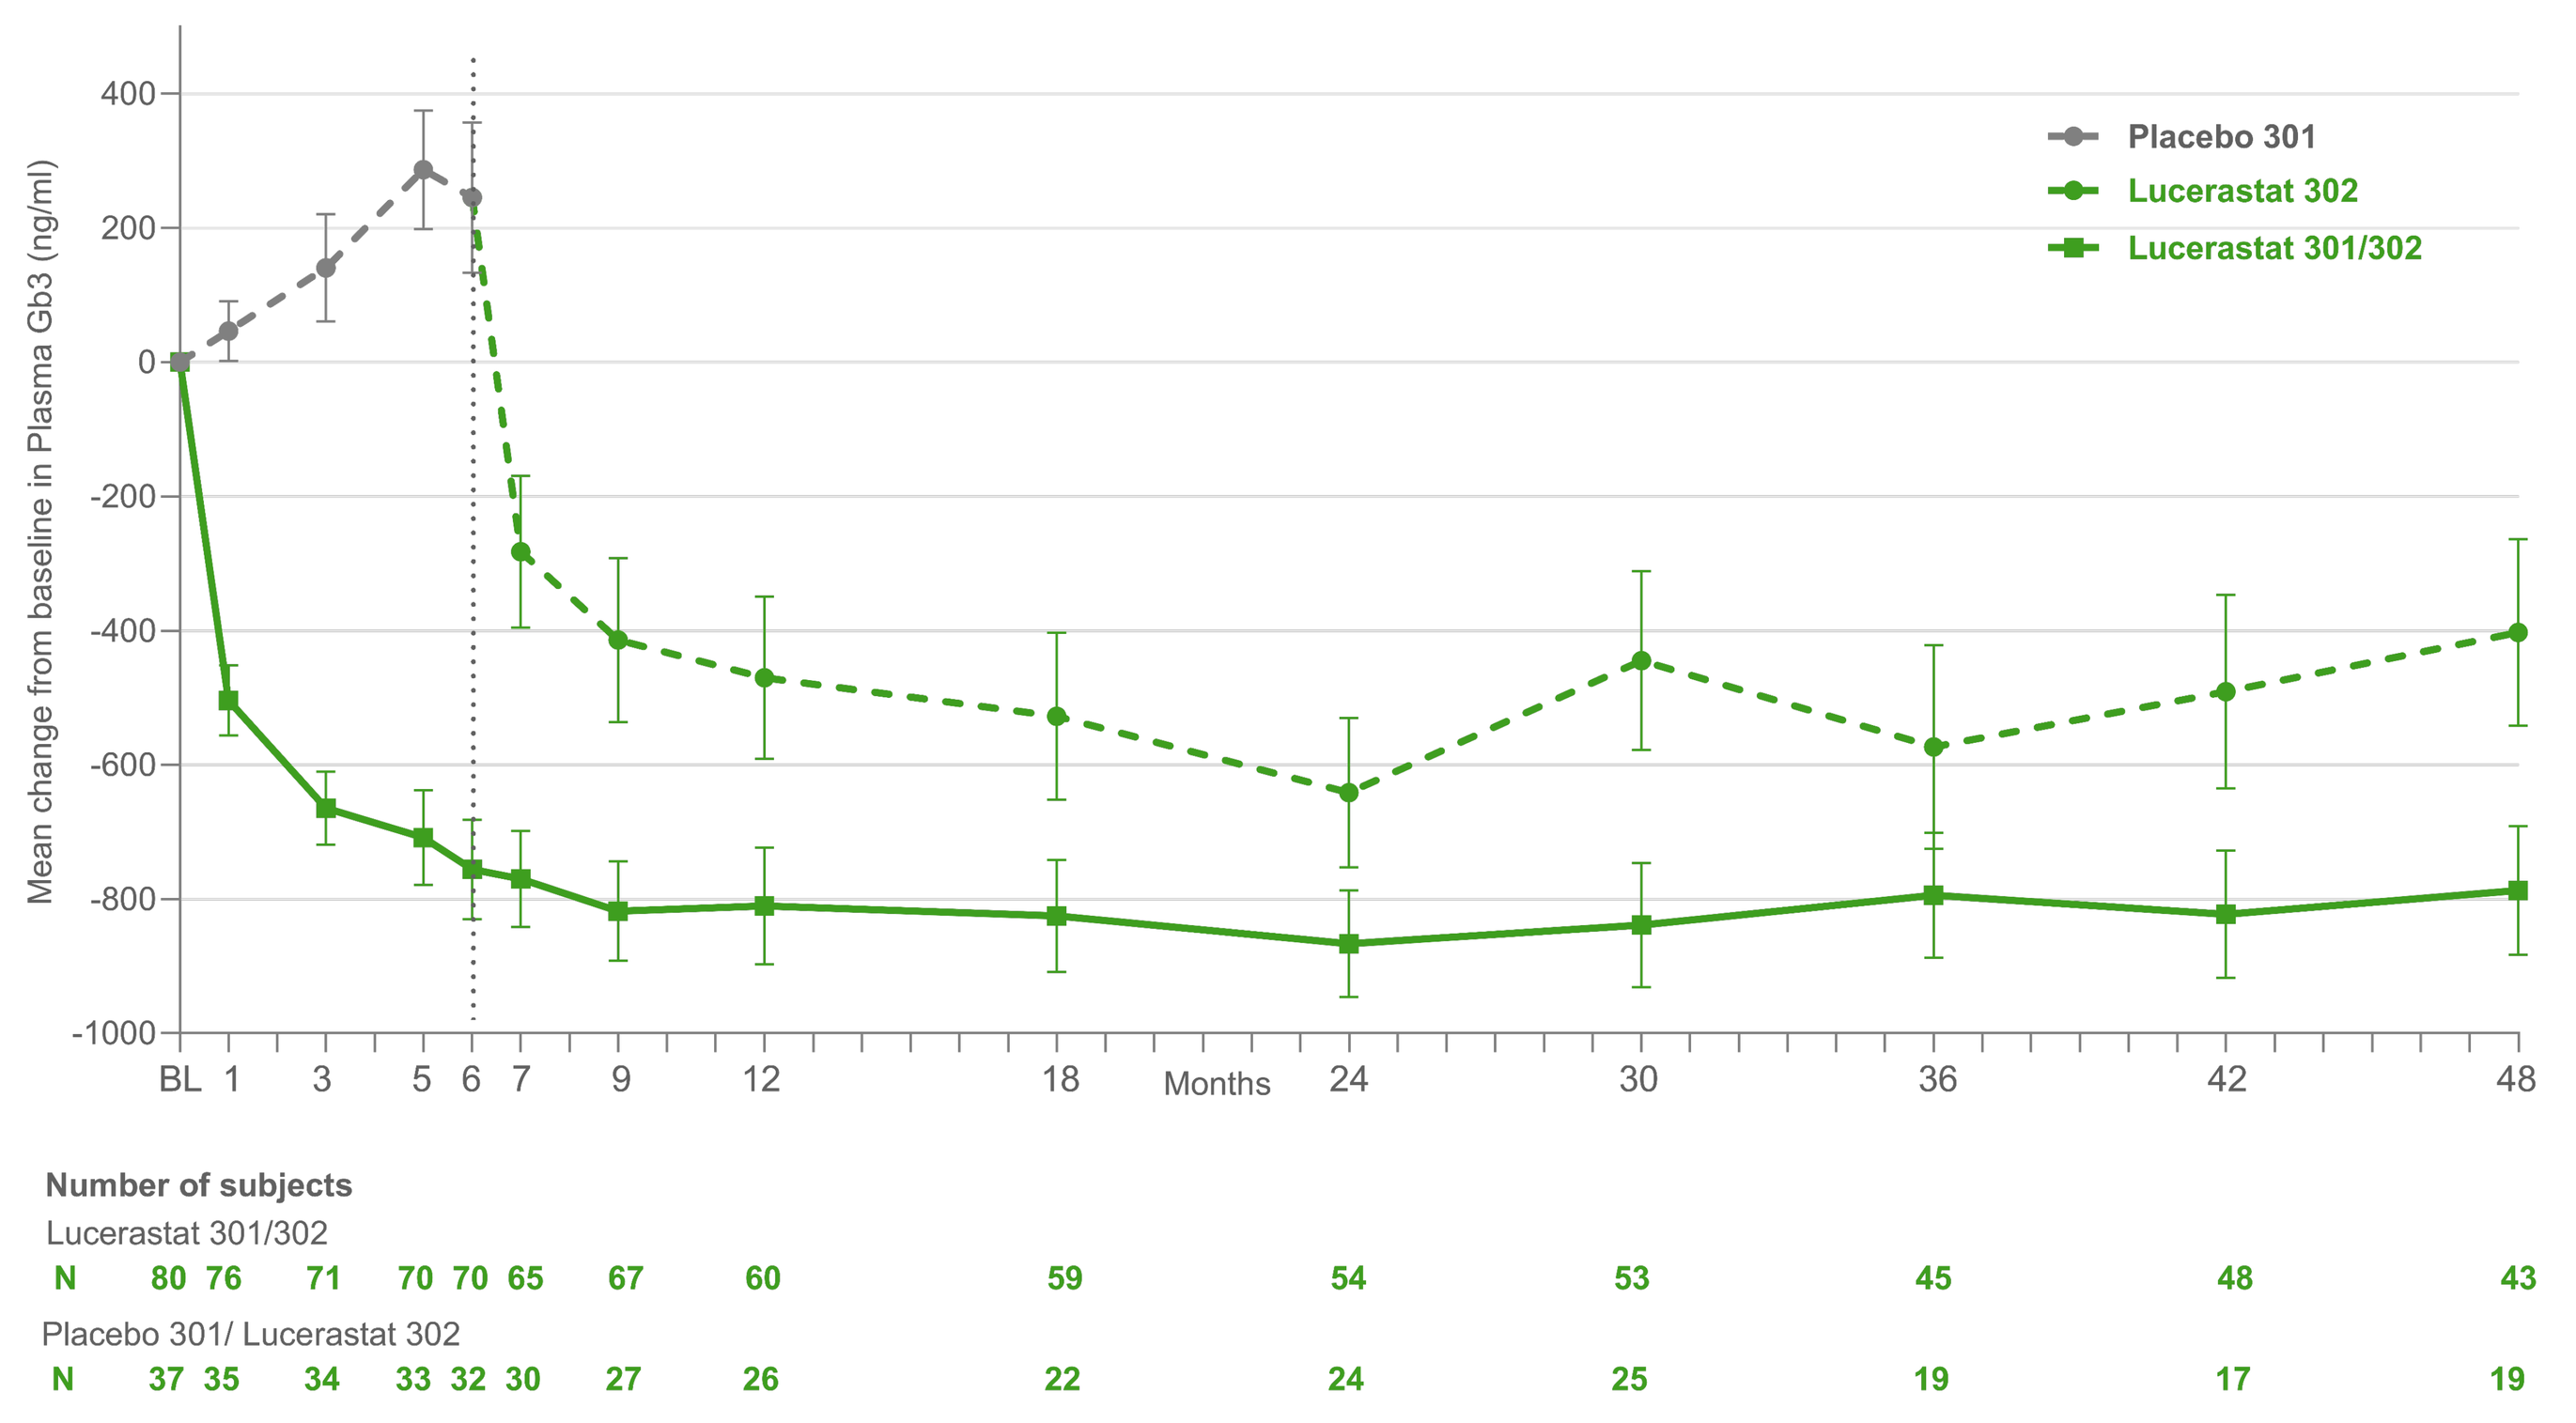
Mean change from baseline in Plasma Gb3 (ng/ml)
400
200
0
-200
-400
-600
-800
-1000
BL 1 3 5 6 7 9 12 18 Months 24 30 36 42 48
Placebo 301
Lucerastat 302
Lucerastat 301/302
Number of subjects
Lucerastat 301/302
N 80 76 71 70 70 65 67 60 59 54 53 45 48 43
Placebo 301/ Lucerastat 302
N 37 35 34 33 32 30 27 26 22 24 25 19 17 19

- Lucerastat led to a sustained 51% decrease in urine Gb3 levels† by Month 30 (urine biomarkers were only measured up to Month 30)

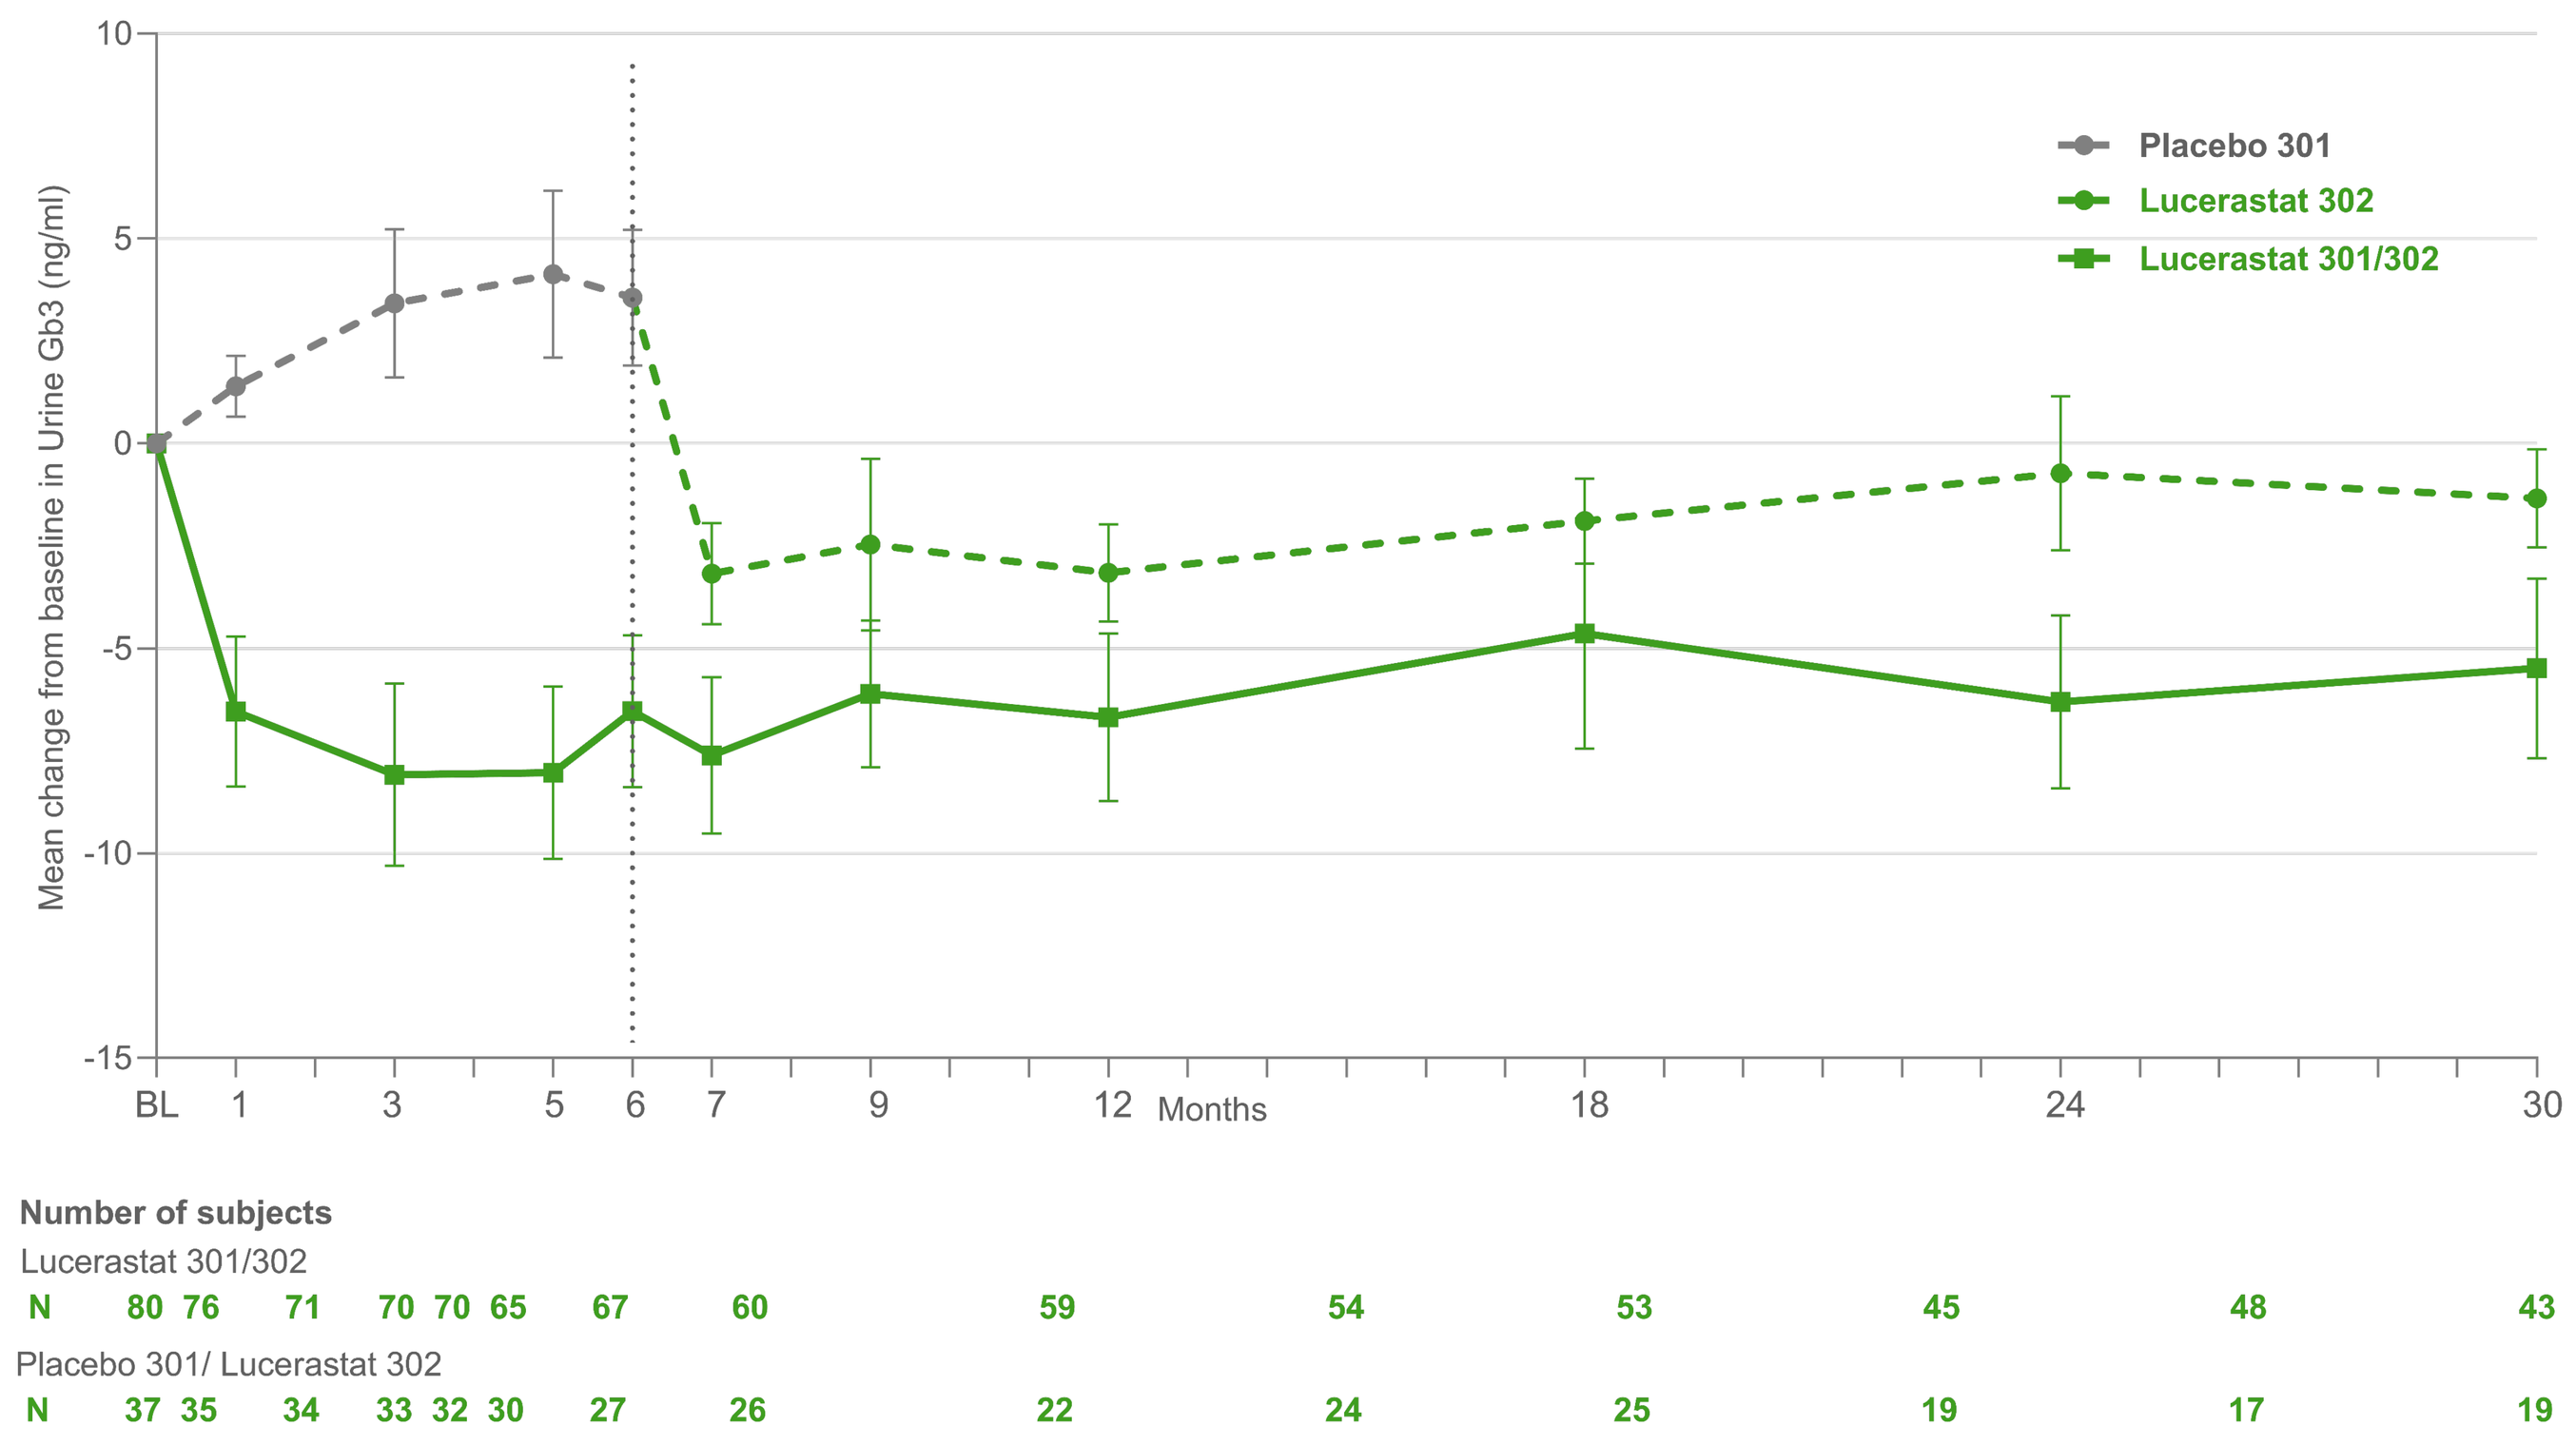
Mean change from baseline in Urine Gb3 (ng/ml)
10
5
0
-5
-10
-15
BL 1 3 5 6 7 9 12 Months 18 24 30
Placebo 301
Lucerastat 302
Lucerastat 301/302
Number of subjects
Lucerastat 301/302
N 80 76 71 70 70 65 67 60 59 54 53 45 48 43
Placebo 301/ Lucerastat 302
N 37 35 34 33 32 30 27 26 22 24 25 19 17 19

Conclusions

Long term exposure to lucerastat was associated with reduced plasma and urine Gb3 levels and slower decline in kidney function compared to pre-randomization, suggesting a renal protective effect, a major therapeutic goal in patients with Fabry disease.

Baseline is the last non-missing value up to and including the date of Study 301 randomization. †*Urine Gb3 levels were normalized by creatinine (µmol Gb3/mol creatinine)*

References

1. Germain, D.P. Orphanet J. Rare Dis. 5, 30 (2010). 2. Morand, O., et al. Adv. Ther. 36, 2866-2880 (2019). 3. Arends, M., et al. J Inherit Metab Dis 41: 141-49 (2018). 4. Wanner, C., et al. Mol. Genet. Metab. Rep. 31, 100862 (2022). 5. Nordbeck, P. et al. Nat. Commun. (2026)

Acknowledgements

Presented at the 22nd Annual World Symposium 2026; February 2-6, 2026; San Diego, USA. This study was funded by Idorsia Pharmaceuticals Ltd.